Effect of combination latency reversing agents and bNAb in SHIV-infected rhesus macaques on antiretroviral therapy

Antonio Solis-Leal[1], Nongthombam Boby[1], Suvadip Mallick[1^], Anna Goodroe[1], Jasmine Kaur[2], Fei Wu[1^^], Grey De La Torre[1], Christine Fennessey[3], Yanhui Cai[2], Jeff Murry[2], Bradon Keele[3], Binhua Ling[1]

[1]Host-Pathogen Interactions Program, Texas Biomedical Research Institute, San Antonio, TX, USA
[2]Gilead Sciences, Inc., Foster City, California, USA
[3]AIDS and Cancer Virus Program, Frederick National Laboratory, Frederick, MD, USA
^current address: Division of Infectious Diseases, Perelman School of Medicine, University of Pennsylvania, PA, USA
^^current address: Buck Institute for Research on Aging, Novato, CA, USA

BACKGROUND

The combination of latency reversing agents (LRA), together with antiretroviral therapy (ART), known as the "Shock and Kill" strategy, is not able to clear established HIV reservoirs. These reservoirs include cells from the myeloid and lymphoid lineage that express the CD4 receptor, and are located throughout the body[1]. This study aims to assess the impact of an LRA with a broadly neutralizing antibody (bNAb) on tissue reservoirs in animals on long-term ART and on the prevention of viral rebound upon ART interruption (ATI).

METHODS

Eight Chinese rhesus macaques were infected with the barcoded virus SHIVAD8E0M[2] and underwent ART starting at 4 weeks post-infection (wpi). These ART-suppressed animals were divided into 2 groups at 49 wpi. The active arm was dosed with AZD5582, then Ciapavir prior to a 6-week ATI (ATI-1) at 80 wpi with a dose of anti-CD8 antibodies to facilitate viral rebound. At 86 wpi, ART was re-initiated with a combination of AZD5582 and PGT121. The control group received daily ART only. At 92 wpi, animals from both groups entered a 2nd ATI (ATI-2) for up to 9 weeks before necropsy (**Figure 1A**). Viral nested PCR, Q-RT-PCR, intact proviral DNA assay (IPDA), immunofluorescence analysis, and RNAscope were used to evaluate the reservoir size and the potential rebounding sources from different cell subsets along different tissues. ELISA test was used to determine levels of PGT121 concentration in blood.

RESULTS

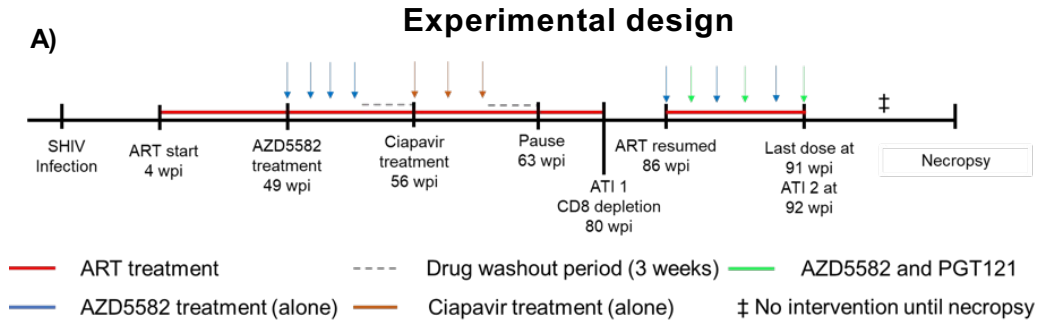
A)
Experimental design
SHIV Infection
ART start 4 wpi
AZD5582 treatment 49 wpi
Ciapavir treatment 56 wpi
Pause 63 wpi
ATI 1 CD8 depletion 80 wpi
ART resumed 86 wpi
Last dose at 91 wpi ATI 2 at 92 wpi
Necropsy
ART treatment
AZD5582 treatment (alone)
Drug washout period (3 weeks)
Ciapavir treatment (alone)
AZD5582 and PGT121
‡ No intervention until necropsy

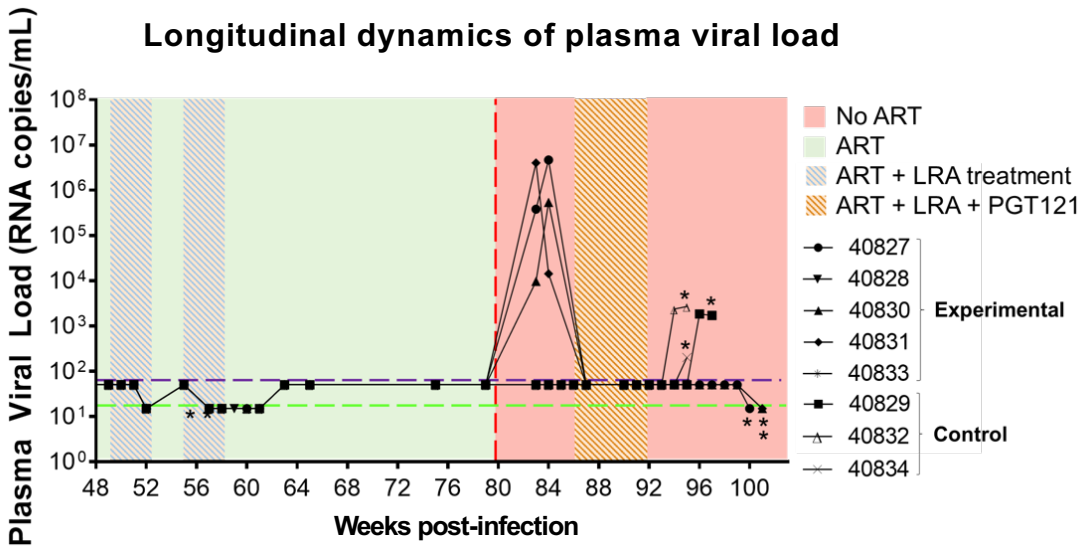
B)
Longitudinal dynamics of plasma viral load
Plasma Viral Load (RNA copies/mL)
Weeks post-infection
No ART
ART
ART + LRA treatment
ART + LRA + PGT121
40827
40828
40830
40831
40833
Experimental
40829
40832
40834
Control

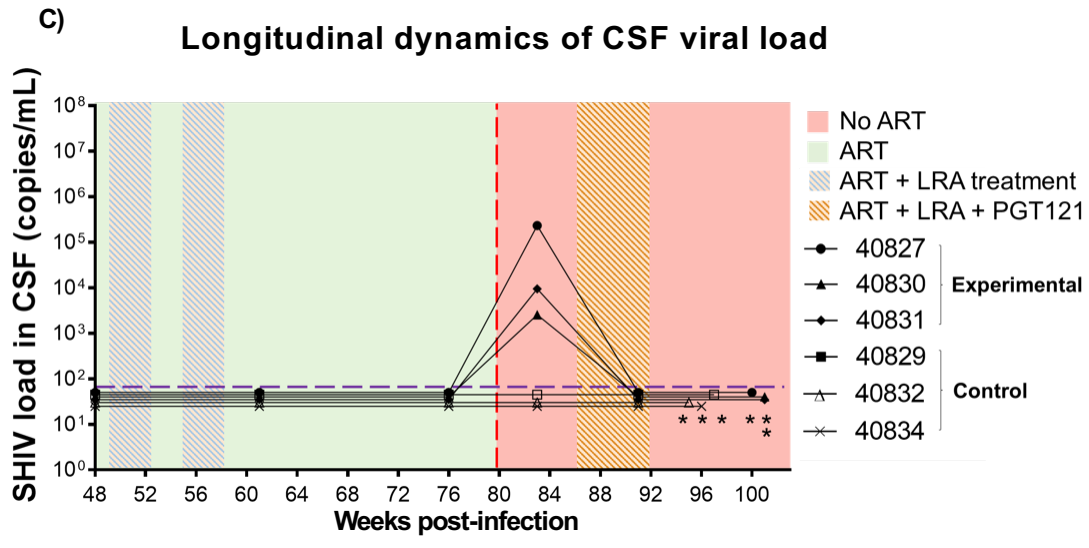
C)
Longitudinal dynamics of CSF viral load
SHIV load in CSF (copies/mL)
Weeks post-infection
No ART
ART
ART + LRA treatment
ART + LRA + PGT121
40827
40830
40831
Experimental
40829
40832
40834
Control

Figure 1: A) Experimental design **B)** Dynamics of plasma viral load. **C)** Dynamics of CSF viral load. Vertical dashed red line represent anti-CD8 antibody infusion. Purple and green horizontal dashed lines represent the limit of detection for the quantification of SHIV of 81 copies/mL and 22 copies/mL respectively. * marks the necropsy of the animal.

PVL maintained < 81 copies/ml while on ART. During the ATI-1 period, experimental animals experienced a rebound that was detected both in PVL and CSF (**Figure 1B and C**). Viral rebound was not detected in experimental animals during ATI-2 neither in plasma or CSF, while control animals experienced a rebound detected in plasma. Further SHIV detection analysis in tissues at different timepoints and necropsy revealed comparable viral detection by different methods (**Figure 2**). Single LRA-treatment with AZD5582 first, followed by Ciapavir while under ART seemed to induce a significant decrease in the intact proviral reservoir found in axillary lymph nodes compared to the control group which only received ART. In addition, the control group showed a mean of 32 intact provirus/10^6 cells in inguinal lymph nodes at necropsy, but none was detected in the experimental animals. Moreover, SHIV was detected in multiple regions of the brain, being readily detectable in basal ganglia, hippocampus and hypothalamus (**Figure 3A**). Further viral quantification revealed low concentrations in most regions, while still detectable for some animals in temporal lobe, hypothalamus and thalamus (**Figure 3B**). In addition, some intact proviral levels ranging between 7.5 and 30.4 intact provirus/10^6 cells were detected in temporal and parietal lobe, hippocampus, and hypothalamus in some of the animals from both groups (**Figure 3C**).

Table 1. SHIV detection in multiple tissues

A)

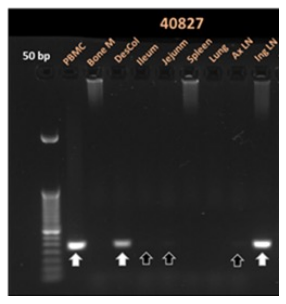
40827
50 bp
PBMC
Bone M
DesCol
Ileum
Jejunum
Spleen
Lung
Ax LN
Ing LN

		PBMC	BoneM	Colon	Ileum	Jejunum	Spleen	Lung	AxLN	ColLN	IngLN	MesLN	Brain	Positive %	
Control	40829	+	+	+	n/a	+	+	-	-	+	-	+	+	75%	68%
	40832	-	-	+	+	+	+	-	+	+	+	+	-	63%	
	40834	+	-	+	+	+	+	-	-	+	+	+	+	75%	
LRA treated	40827	+	-	+	+	+	-	-	-	n/a	+	+	+	64%	65%
	40830	-	+	+	+	-	-	+	+	-	+	-	+	58%	
	40831	-	-	-	+	+	+	+	+	+	n/a	+	+	73%	
Positive %		50%	33%	83%	100%	83%	66%	33%	50%	80%	80%	83%	83%		

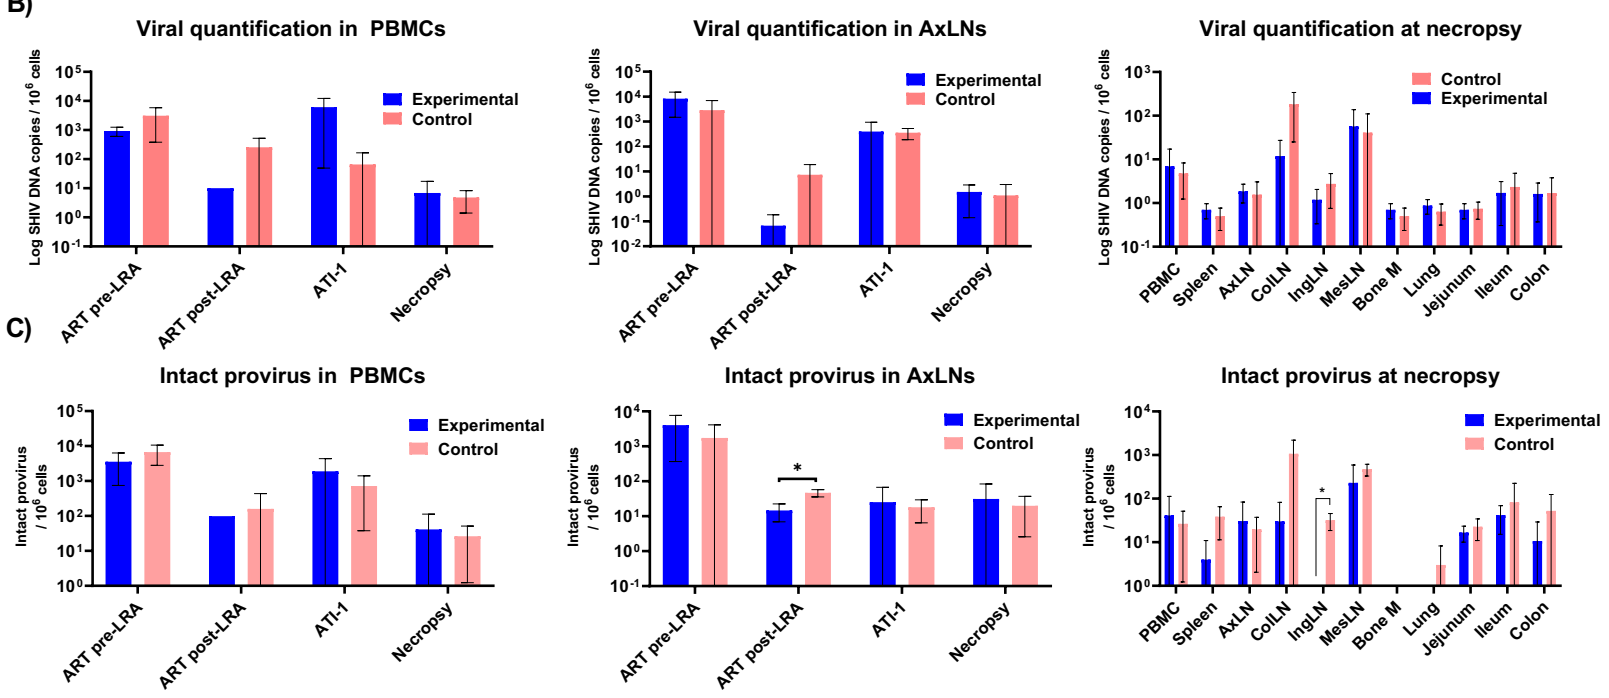
B)
Viral quantification in PBMCs
Viral quantification in AxLNs
Viral quantification at necropsy
Log SHIV DNA copies / 10^6 cells
Experimental
Control
ART pre-LRA
ART post-LRA
ATI-1
Necropsy
PBMC
Spleen
AxLN
ColLN
IngLN
MesLN
Bone M
Lung
Jejunum
Ileum
Colon
C)
Intact provirus in PBMCs
Intact provirus in AxLNs
Intact provirus at necropsy
Intact provirus / 10^6 cells

Figure 2: A) Viral detection in tissues. **B)** Cell-associated viral DNA load in tissues. **C)** Level of intact provirus in tissues. * $p < 0.05$.

Table 2. Comparative SHIV detection in different brain regions

A)

		Frontal lobe	Temporal lobe	Parietal lobe	Basal ganglia	Brain stem	Hippo campus	Hypo thalamus	Thalamus	Positive %	
Control	40829	+	+	+	+	+	+	+	-	88%	84%
	40832	+	-	+	+	+	+	+	-	75%	
	40834	+	+	+	+	+	+	+	-	88%	
LRA Treated	40827	+	+	+	+	-	+	+	+	88%	80%
	40830	-	+	+	+	+	+	+	+	88%	
	40831	+	-	-	+	+	+	+	-	63%	
Positive %		83%	67%	83%	100%	83%	100%	100%	33%		

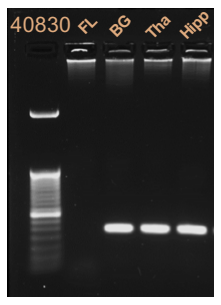
40830
FL
BG
Tha
Hipp

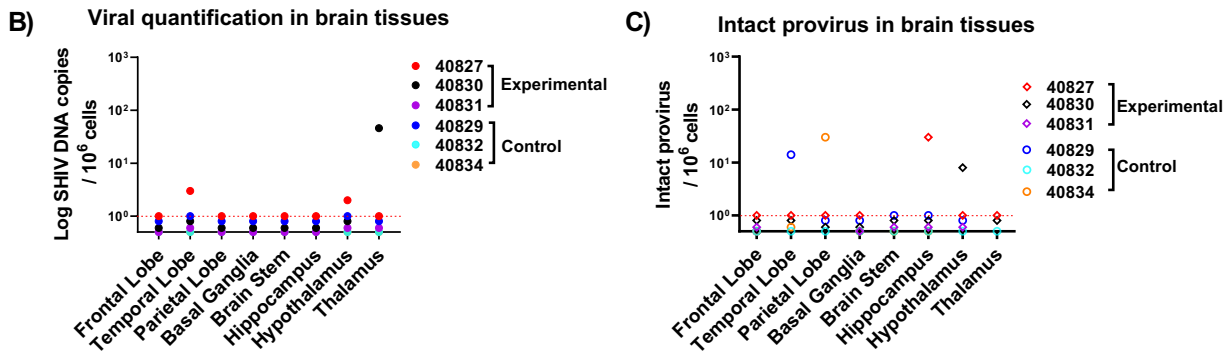
B)
Viral quantification in brain tissues
Log SHIV DNA copies / 10^6 cells
40827
40830
40831
Experimental
40829
40832
40834
Control
Frontal Lobe
Temporal Lobe
Parietal Lobe
Basal Ganglia
Brain Stem
Hippocampus
Hypothalamus
Thalamus
C)
Intact provirus in brain tissues
Intact provirus / 10^6 cells

Figure 3: A) Viral detection in brain tissues. **B)** Viral quantification in brain tissues. **C)** Intact provirus in brain tissues. Red dashed lines represent the limit of detection of the assay of 1 intact provirus per 10^6 cells.

vRNA detection in brain tissues

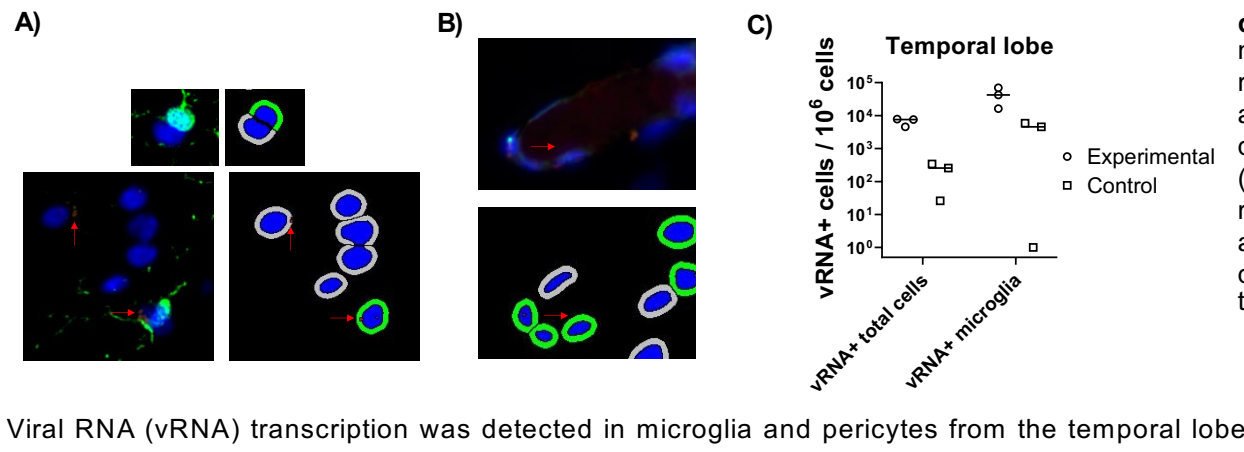
A)
B)
C)
Temporal lobe
vRNA+ cells / 10^6 cells
Experimental
Control
vRNA+ total cells
vRNA+ microglia

Figure 4: vRNA transcription quantification A) HALO analysis of microglia (Iba-1). Left panels are representative IF images. Right panels are representative viral and cellular detection**. B)** HALO analysis of pericytes (PDGFR-β). Top panel is a representative IF image. Bottom panel is a representative viral and cellular detection**. C)** vRNA quantification in temporal lobe in microglia and total cells.

Viral RNA (vRNA) transcription was detected in microglia and pericytes from the temporal lobe (**Figure 4A and B**). The analysis of the microglia and the total cell population suggested higher levels of viral transcription in the experimental group in this lobe.

PGT121 concentration levels may have prevented a systemic viral rebound in the experimental group at necropsy (**Figure 5A**). In addition, ADA analysis suggested that PGT121 treatment did not induce an immune response in the experimental group (**Figure 5B**).

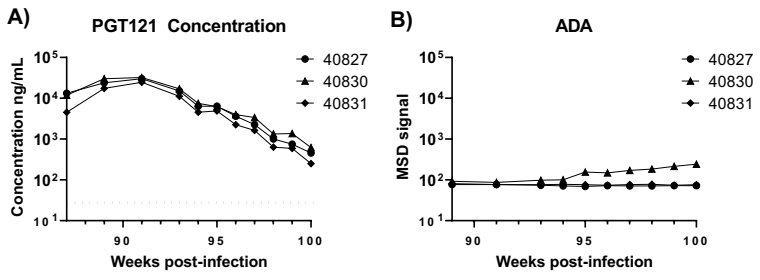
A)
PGT121 Concentration
Concentration ng/mL
Weeks post-infection
40827
40830
40831
B)
ADA
MSD signal
Weeks post-infection
40827
40830
40831

Figure 5 A) PGT121 concentration in blood. **B)** Anti-drug antibodies (ADA) analysis.

CONCLUSIONS

The sustained viral suppression observed in the experimental group suggests that treatment with activators of the non-canonical NF-kB pathway, such as AZD5582, together with ART and PGT121 may decrease LN reservoirs and provide sustained viral control. Further testing in people living with HIV may provide additional evidence of the effectiveness of this strategy upon the need of an ATI. These data also suggest that intact viruses in certain brain regions may serve as HIV reservoirs that could be challenging for eradication, specially, if they are found in different cell types, including microglia or other cell types. Future HIV cure strategies may need to include interventions that specifically target these resilient and varied reservoirs.

REFERENCES

1 Deeks, S. G. et al. Research priorities for an HIV cure: International AIDS Society Global Scientific Strategy 2021. Nat Med 27, 2085-2098.
2 Khanal S. et al. In Vivo Validation of the Viral Barcoding of Simian Immunodeficiency Virus SIVmac239 and the Development of New Barcoded SIV and Subtype B and C Simian-Human Immunodeficiency Viruses. 2019. J Virol 94(1): e01420-19.

Presented at IAS 2023, the 12th IAS Conference on HIV Science

IAS 2023
23 – 26 July