Recommendations for Missed Oral Lenacapavir Loading Doses Using Population-Pharmacokinetics-Based Simulations

TUPEB14

Scan for more information

Renu Singh,[1] Naveed A. Shaik,[1] Francesco Bellanti,[2] Craig Comisar,[2] Kishore Polireddy,[2] Martin Rhee,[1] Ramesh Palaparthy[1]

[1]Gilead Sciences, Inc., Foster City, CA, USA; [2]Certara, Inc., Princeton, NJ, USA

Key Findings

- Lenacapavir (LEN) is approved for treating multidrug-resistant HIV-1, in combination with other antiretrovirals, for heavily treatment-experienced people with HIV (PWH)
- The dosing regimen used in the pivotal Phase 2/3 study (CAPELLA) is oral LEN loading doses (Days 1 and 2: 600 mg; Day 8: 300 mg) then subcutaneous (SC) LEN dosing (927 mg every 6 months [Q6M] starting on Day 15) (Phase 2/3 regimen)
- LEN has a long terminal/apparent half-life of 10–12 days (oral) and 8–12 weeks (SC)
- Oral LEN loading is critical to achieving and maintaining efficacious concentration. Therefore, it is important to characterize options to manage missed oral doses
- Various scenarios were simulated to guide missed dose recommendations for oral loading doses that maintain LEN concentrations above the therapeutic target
- If the Day 2 oral dose is missed:
 - by <6 days, a 600 mg oral dose should be taken as soon as possible, and 300 mg on Day 8
 - by ≥6 days, a 600 mg oral dose should be taken as soon as possible, and a 300 mg on Day 15
- If the Day 8 oral dose is missed:
 - by <6 days, a 300 mg oral dose should be taken as soon as possible
 - by ≥6 days, a 300 mg oral dose should be taken on Day 15

Conclusions

- Given the long half-life of oral LEN, the dosing window for LEN oral loading is wide and forgiving when a loading dose is missed
- LEN oral loading ensures consistent therapeutic concentrations
- The recommended options for LEN oral loading doses to manage a missed oral loading dose are shown in **Table 1**

Table 1. Summary of missed dose recommendations for LEN oral loading in Phase 2/3 regimen (start and restart of treatment)

Loading Dose Day	Missed Dose Scenarios	Recommendation*
Day 2	Day 2 oral dose missed by <6 days	Take 600 mg oral dose as soon as possible, and 300 mg on Day 8
	Day 2 oral dose missed by ≥6 days	Take 600 mg oral dose as soon as possible, and 300 mg on Day 15
Day 8	Day 8 oral dose missed by <6 days	Take 300 mg oral dose as soon as possible
	Day 8 oral dose missed by ≥6 days	Take 300 mg oral dose on Day 15

*In addition to SC 927 mg on Day 15.
LEN, lenacapavir; SC, subcutaneous

Background

- LEN is a potent first-in-class capsid inhibitor, recently approved in heavily treatment-experienced adults for the treatment of multidrug-resistant HIV-1 infection, in combination with other antiretrovirals[1,2]
- LEN has been demonstrated to be efficacious with a favorable safety profile in the pivotal Phase 2/3 CAPELLA study (NCT04150068)[3]
 - Participants received LEN oral loading doses (Days 1 and 2: 600 mg; Day 8: 300 mg) then maintenance with 927 mg LEN SC Q6M, starting on Day 15 (Phase 2/3 regimen)[1,2]
- LEN has a long terminal/apparent half-life of 10–12 days and 8–12 weeks following oral and SC administration, respectively[1,2]
- Current data indicate maximal antiviral activity is achieved at a mean trough concentration of 15.5 ng/mL,[4] i.e., the inhibitory quotient-4 (IQ4; ≥4-fold higher than the *in vitro* protein-adjusted 95% effective concentration in MT-4 cells)[5]
- Since LEN oral loading doses are critical in achieving and maintaining plasma LEN concentrations ≥IQ4, it is important to characterize potential options to manage missed oral loading doses

Objective

- The objective here was to guide missed oral loading dose recommendations for PWH using a population-pharmacokinetic (PopPK) simulation approach

Methods

- A previously developed two-compartment PopPK model with first-order absorption and linear elimination[6] was used to simulate plasma LEN concentrations following various missed oral loading dose scenarios
- **Figure 1** shows simulated plasma LEN concentration–time profiles for the oral loading portion of the Phase 2/3 dosing regimen (taken as scheduled)
- Simulated plasma LEN concentration–time profiles in PWH for different missed-dose scenarios of Day 2 and Day 8 oral loading doses were evaluated at the start of treatment and restart of treatment (when a LEN SC dose was missed by >2 weeks then a restart of treatment was needed from Day 1)
- The following scenarios were simulated:
 - Day 2 oral dose missed by <6 days
 - Day 2 oral dose missed by ≥6 days
 - Day 8 oral dose missed by <6 days
 - Day 8 oral dose missed by ≥6 days
- In all scenarios, LEN SC injection was given on Day 15

Figure 1. Simulated LEN concentration-time profiles in PWH for the oral loading portion of the Phase 2/3 dosing regimen

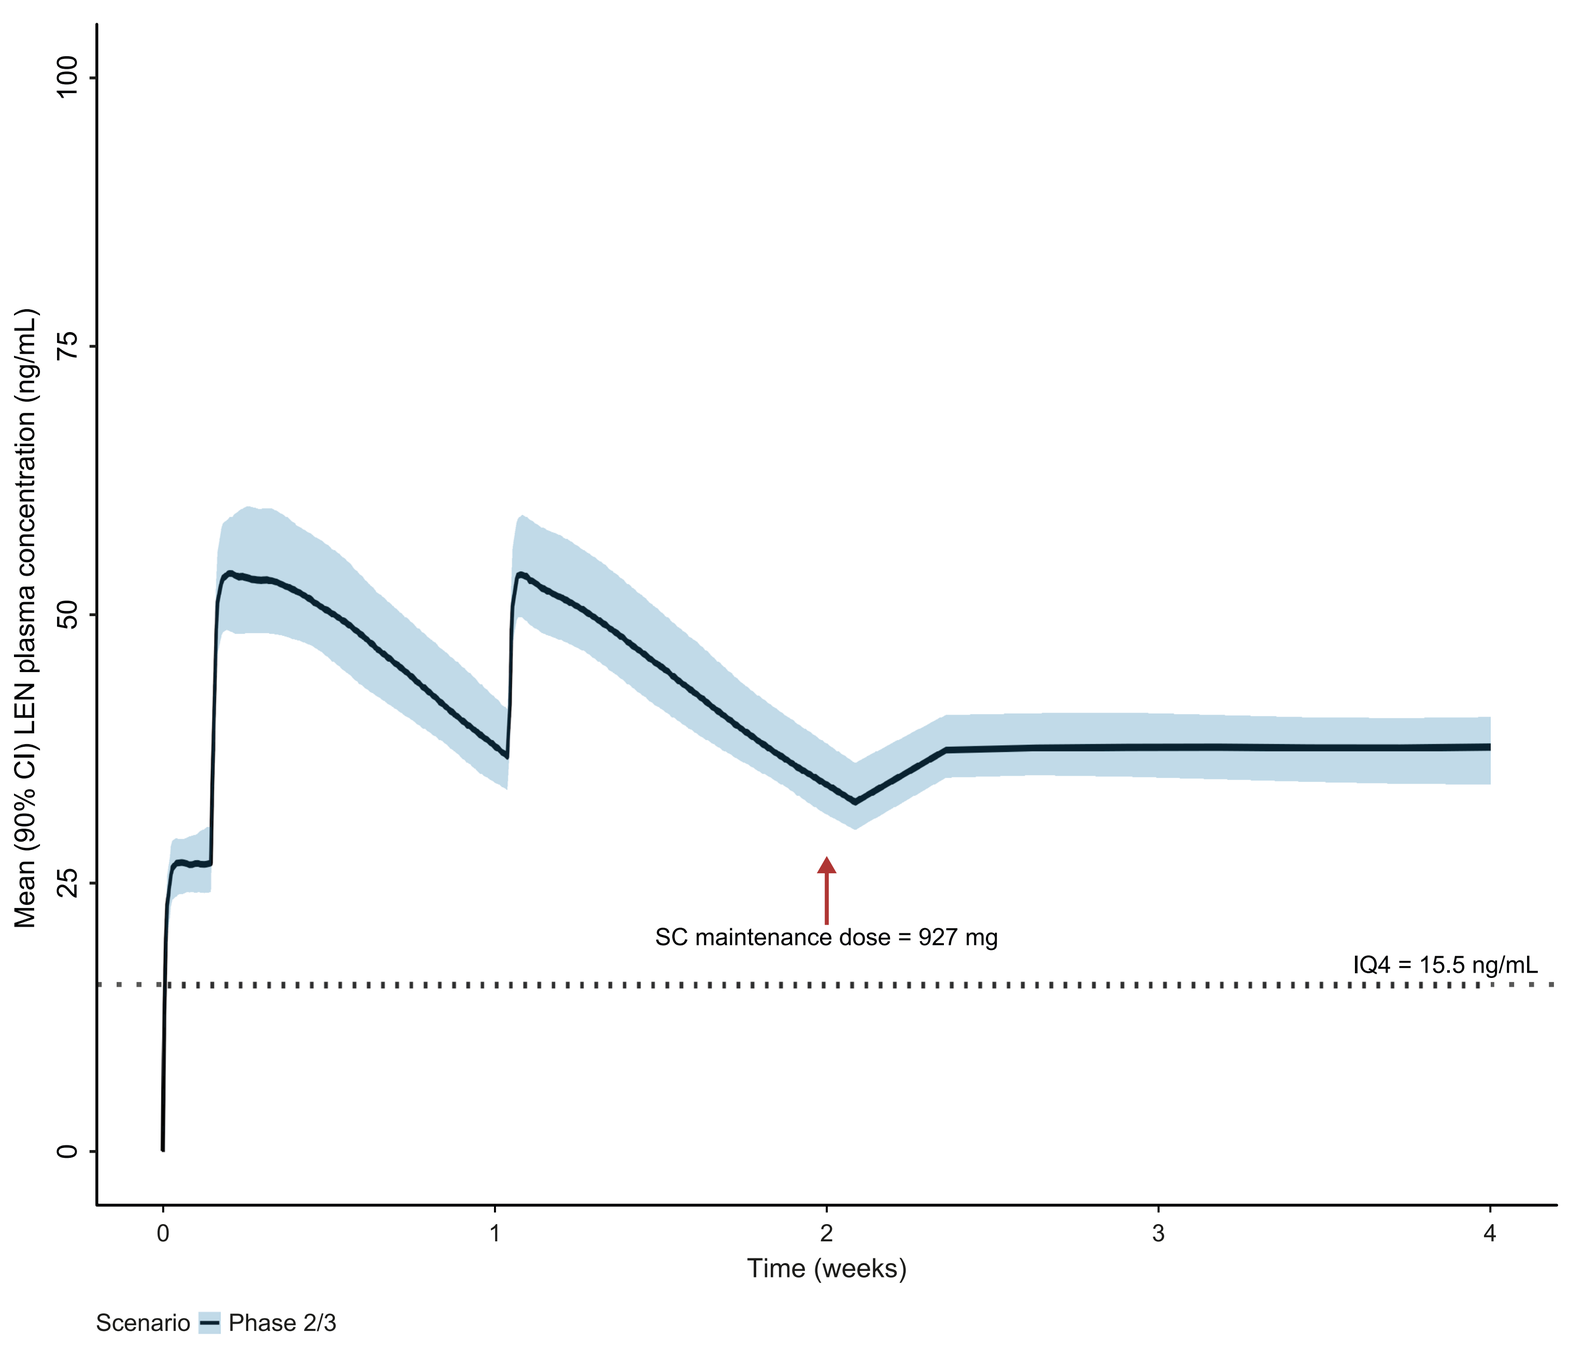
Mean (90% CI) LEN plasma concentration (ng/mL)
SC maintenance dose = 927 mg
IQ4 = 15.5 ng/mL
Time (weeks)
Scenario Phase 2/3

LEN oral loading doses are: 600 mg on Days 1 and 2, and 300 mg on Day 8.
CI, confidence interval; IQ4, inhibitory quotient-4; LEN, lenacapavir; PWH, people with HIV; SC, subcutaneous

Results

Missed dose scenarios at the start of treatment

- Simulated LEN plasma concentrations for each scenario are shown in **Figure 2**
- When the Day 2 oral dose was missed, the mean (90% CI) LEN plasma concentrations were consistently ≥IQ4 if:
 - the 600 mg oral dose was taken as soon as possible, and a 300 mg dose on Day 8 (Day 2 dose missed by <6 days) (**Figure 2A**) OR
 - the 600 mg oral dose (instead of 300 mg) was taken as soon as possible and a 300 mg oral dose on Day 15 along with a 927 mg SC dose (Day 2 dose missed by ≥6 days) (**Figure 2B**)
- When the Day 8 oral dose was missed, therapeutic LEN plasma concentrations (≥IQ4) were maintained if:
 - a 300 mg oral dose was taken as soon as possible (Day 8 dose missed by <6 days) (**Figure 2C**) OR
 - a 300 mg oral dose was taken on Day 15 along with a 927 mg SC dose (Day 8 dose missed by ≥6 days) (**Figure 2D**)
- Missing Day 2 and Day 8 loading doses resulted in LEN concentrations <IQ4

PWH receiving the Phase 2/3 dosing regimen at the restart of treatment

- Simulated LEN plasma concentrations for each scenario are shown in **Figure 3**
- The dosing recommendations for the start of treatment are also appropriate for the restart of treatment

Figure 2. Simulated LEN concentration–time profiles in PWH who received the Phase 2/3 dosing regimen at the start of treatment

A) Day 2 oral dose missed by <6 days

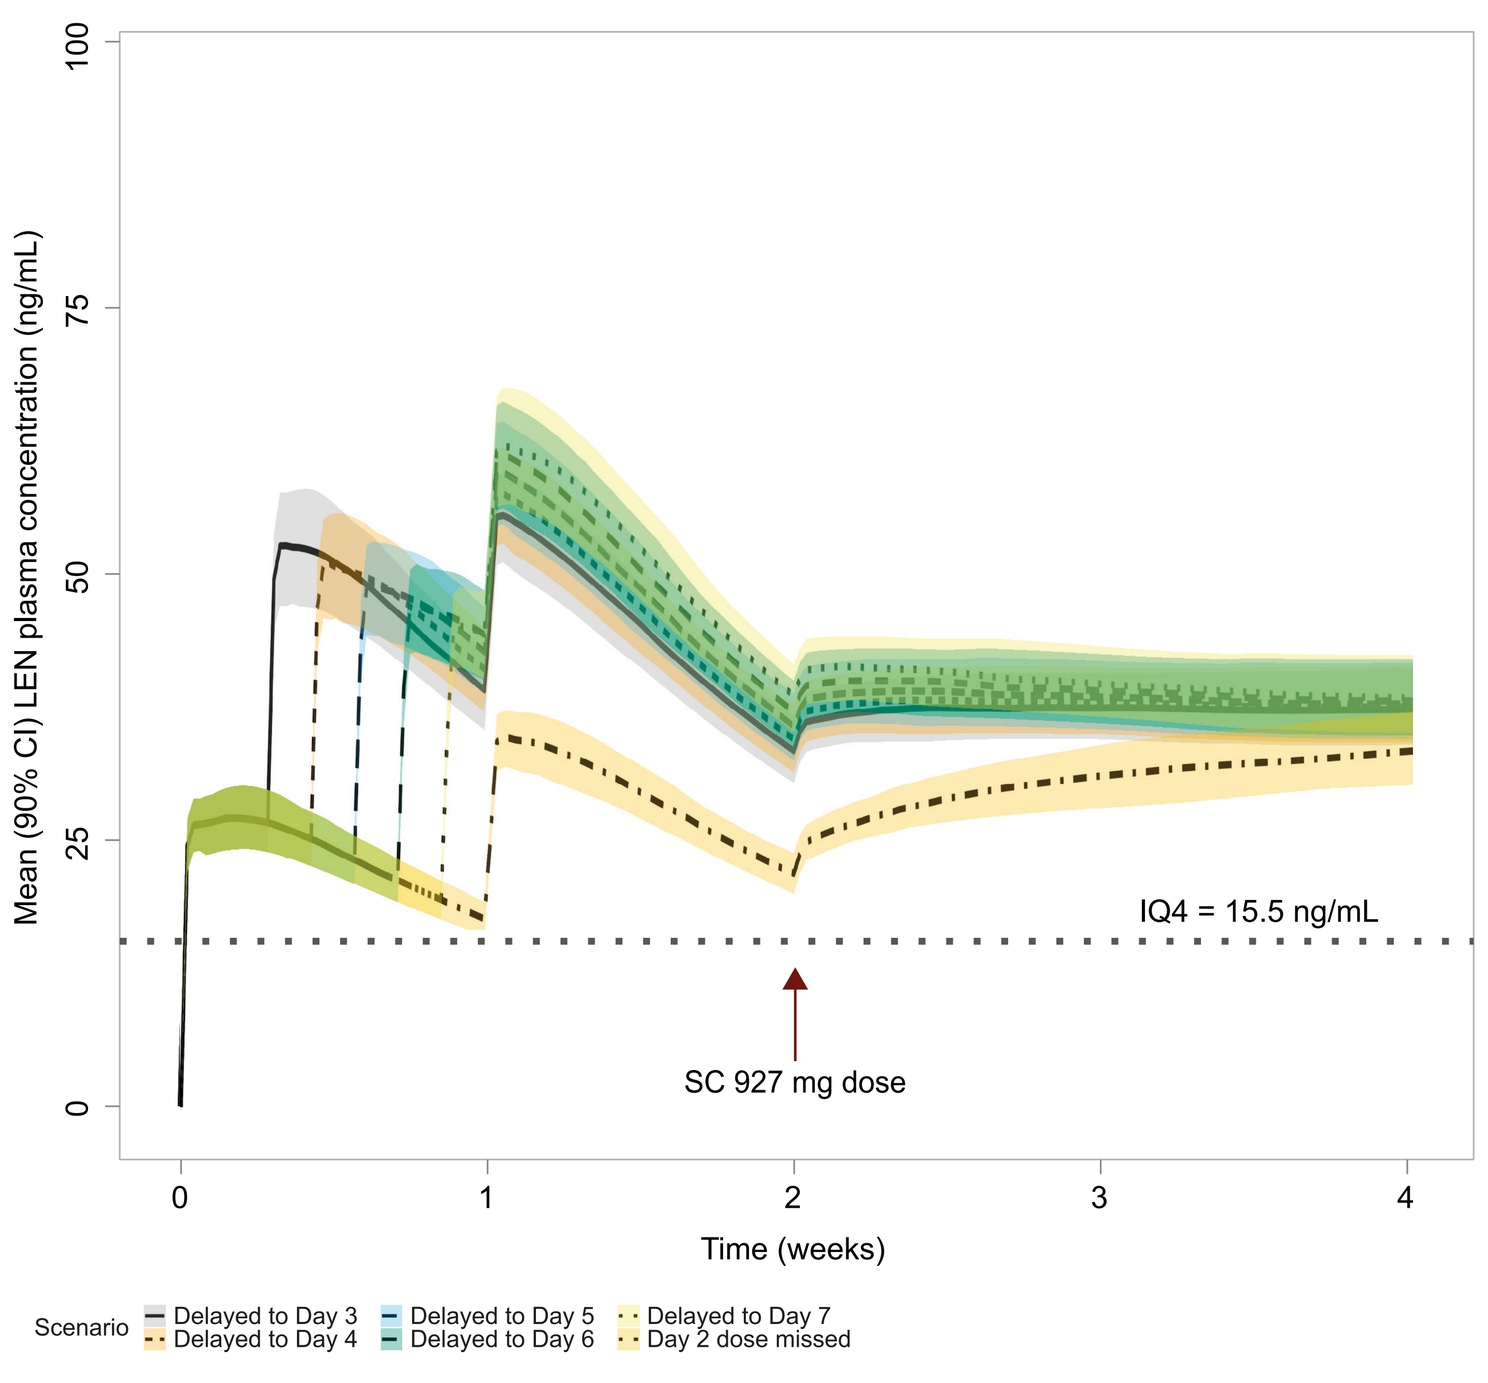
Mean (90% CI) LEN plasma concentration (ng/mL)
IQ4 = 15.5 ng/mL
SC 927 mg dose
Time (weeks)
Scenario
Delayed to Day 3
Delayed to Day 4
Delayed to Day 5
Delayed to Day 6
Delayed to Day 7
Day 2 dose missed

B) Day 2 oral dose missed by ≥6 days

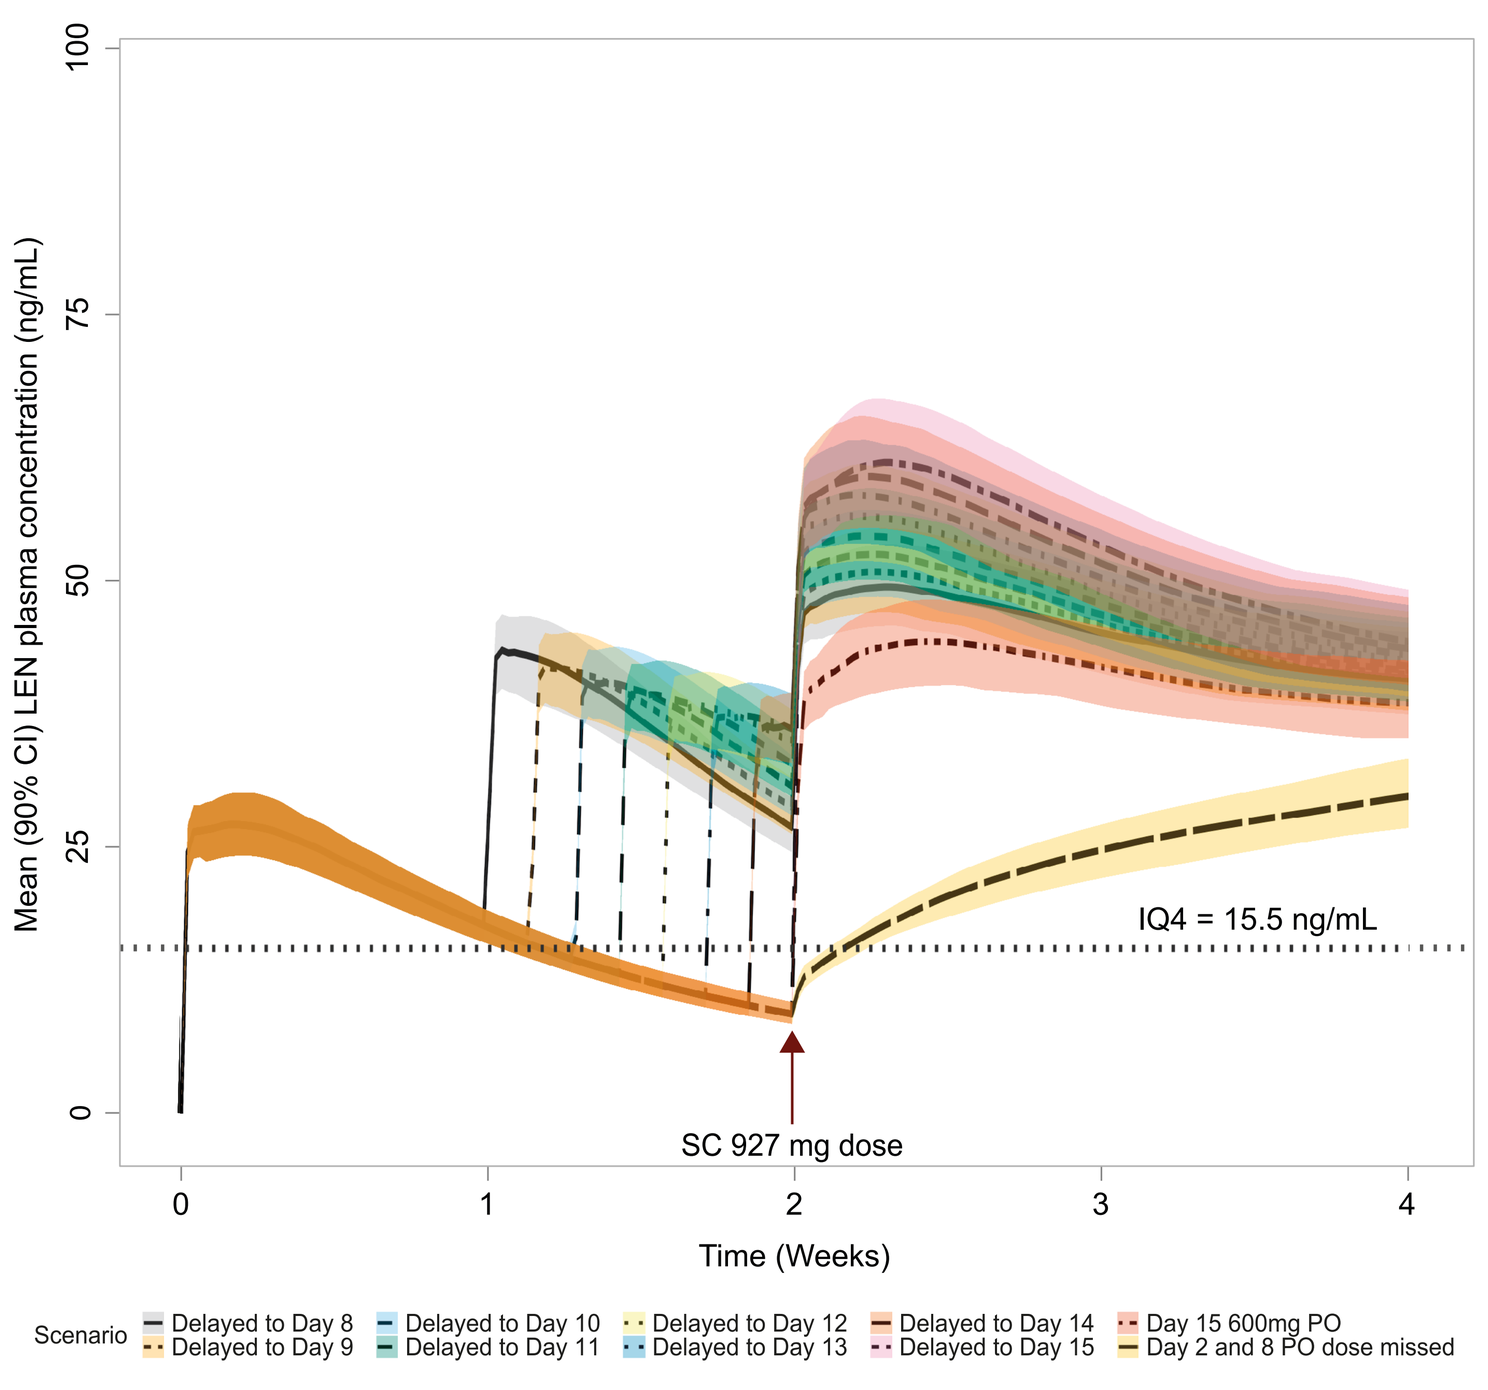
Mean (90% CI) LEN plasma concentration (ng/mL)
IQ4 = 15.5 ng/mL
SC 927 mg dose
Time (Weeks)
Scenario
Delayed to Day 8
Delayed to Day 9
Delayed to Day 10
Delayed to Day 11
Delayed to Day 12
Delayed to Day 13
Delayed to Day 14
Delayed to Day 15
Day 15 600mg PO
Day 2 and 8 PO dose missed

C) Day 8 oral dose missed by <6 days

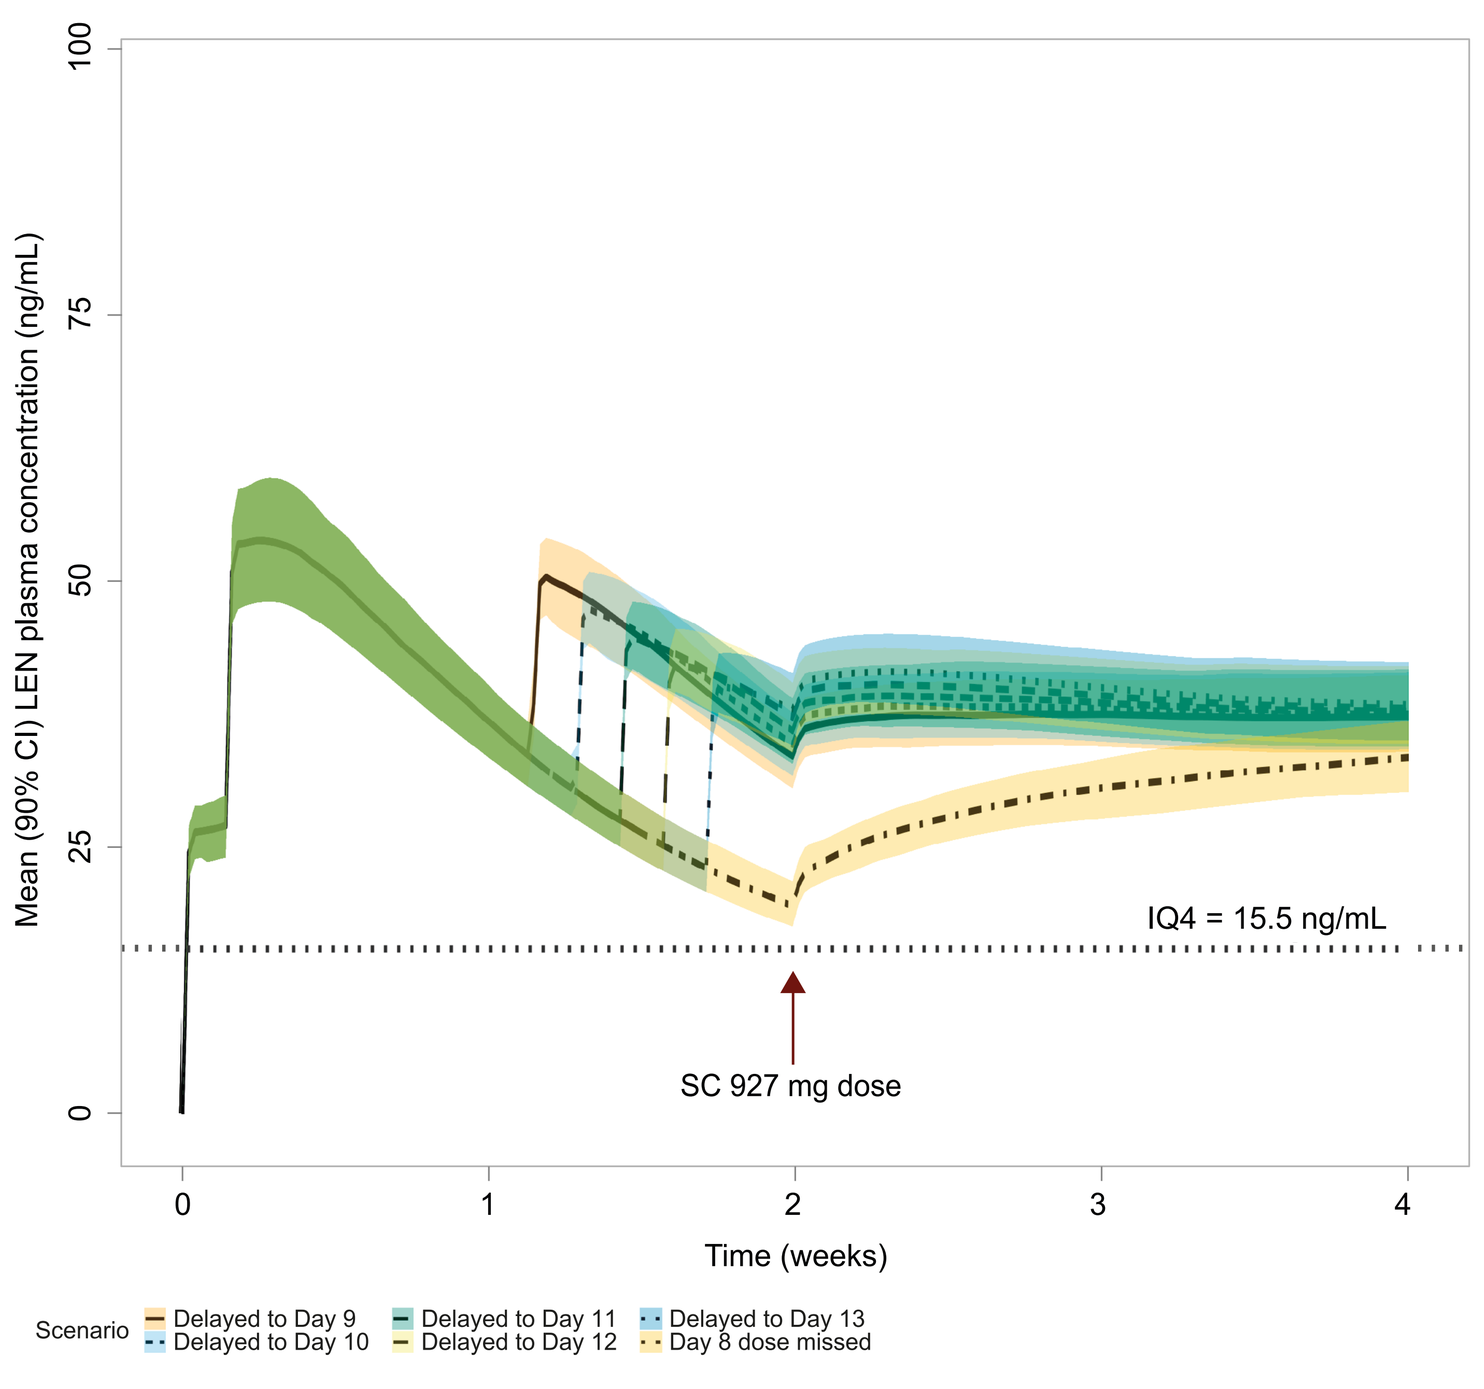
Mean (90% CI) LEN plasma concentration (ng/mL)
IQ4 = 15.5 ng/mL
SC 927 mg dose
Time (weeks)
Scenario
Delayed to Day 9
Delayed to Day 10
Delayed to Day 11
Delayed to Day 12
Delayed to Day 13
Day 8 dose missed

D) Day 8 oral dose missed by ≥6 days

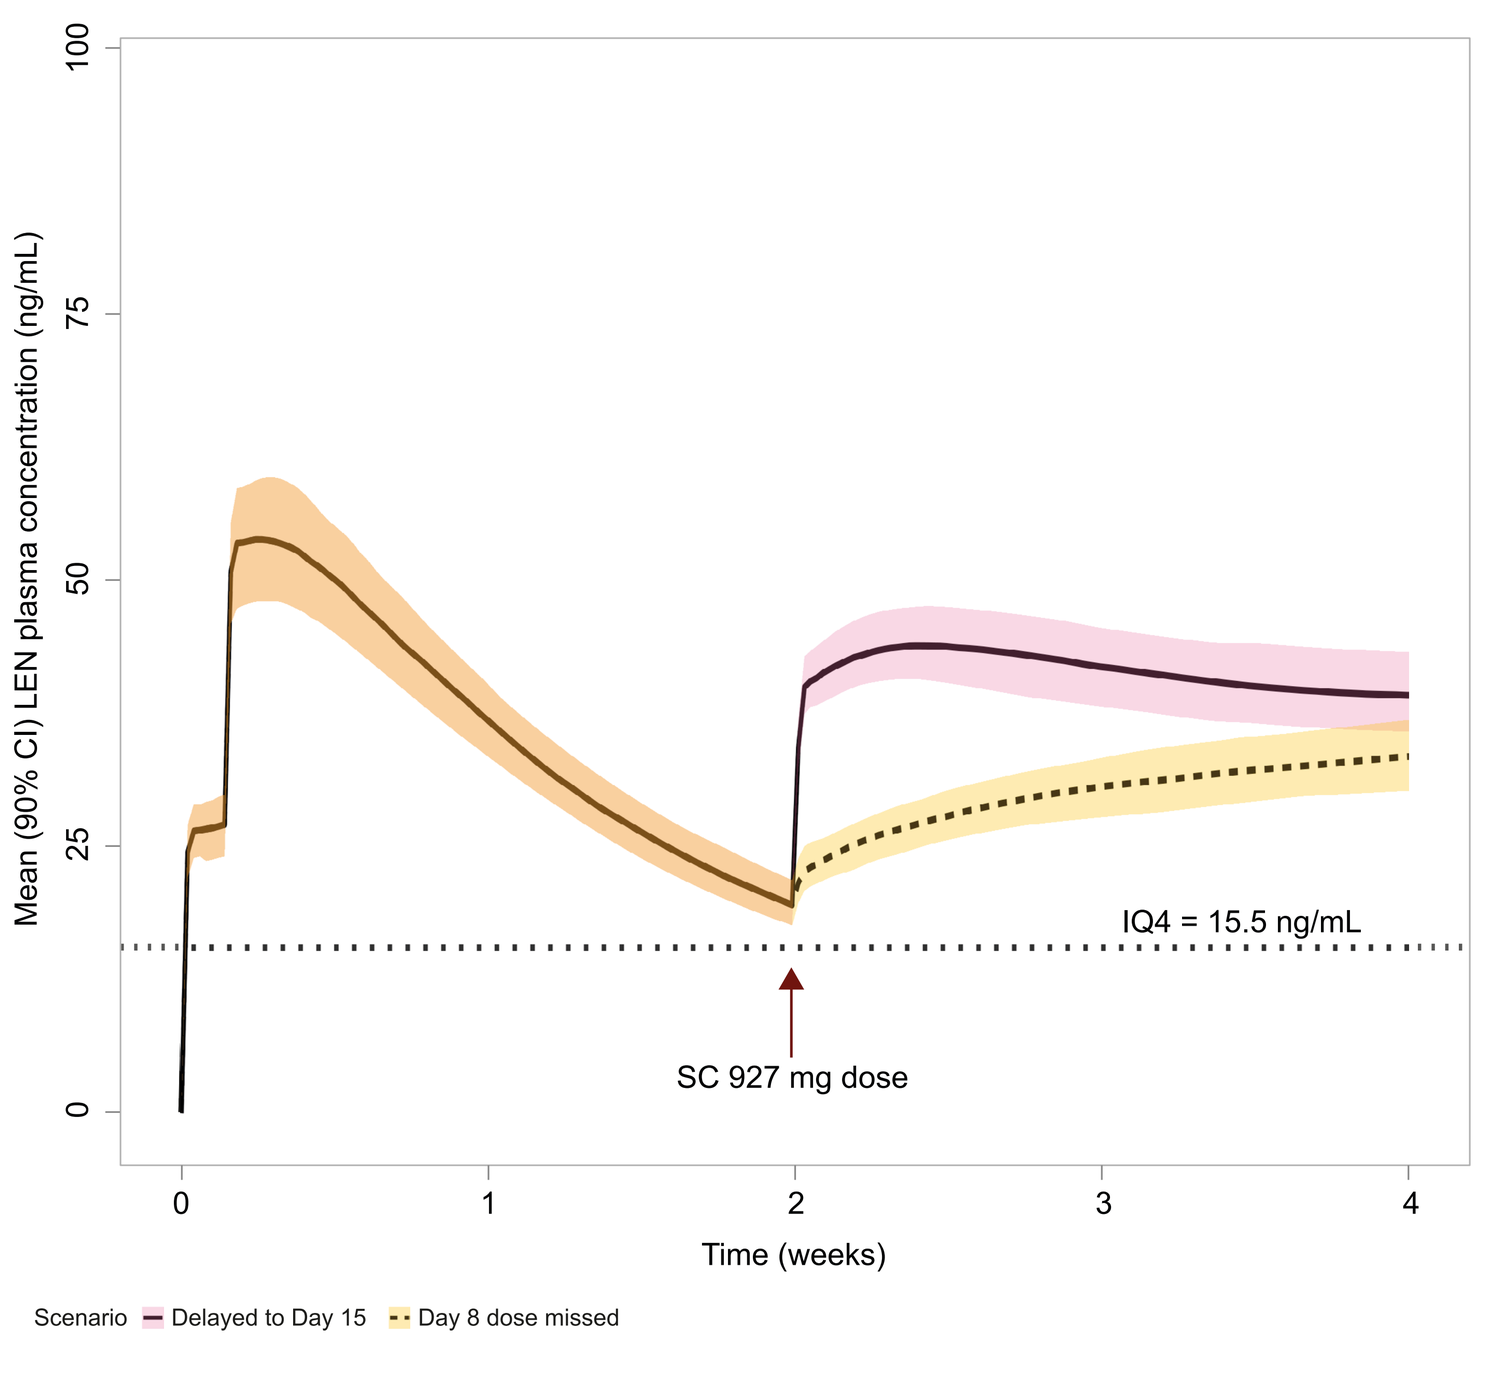
Mean (90% CI) LEN plasma concentration (ng/mL)
IQ4 = 15.5 ng/mL
SC 927 mg dose
Time (weeks)
Scenario
Delayed to Day 15
Day 8 dose missed

Lines represent the simulated mean. Shaded areas represent the 90% confidence interval of the simulated mean.
In all scenarios, LEN SC dose was simulated on Day 15
CI, confidence interval; IQ4, inhibitory quotient-4; LEN, lenacapavir; PO, per os (oral); PWH, people with HIV; SC, subcutaneous

Figure 3. Simulated LEN concentration–time profiles in PWH who received the Phase 2/3 dosing regimen at the restart of treatment at steady state

A) Day 2 oral dose missed by <6 days

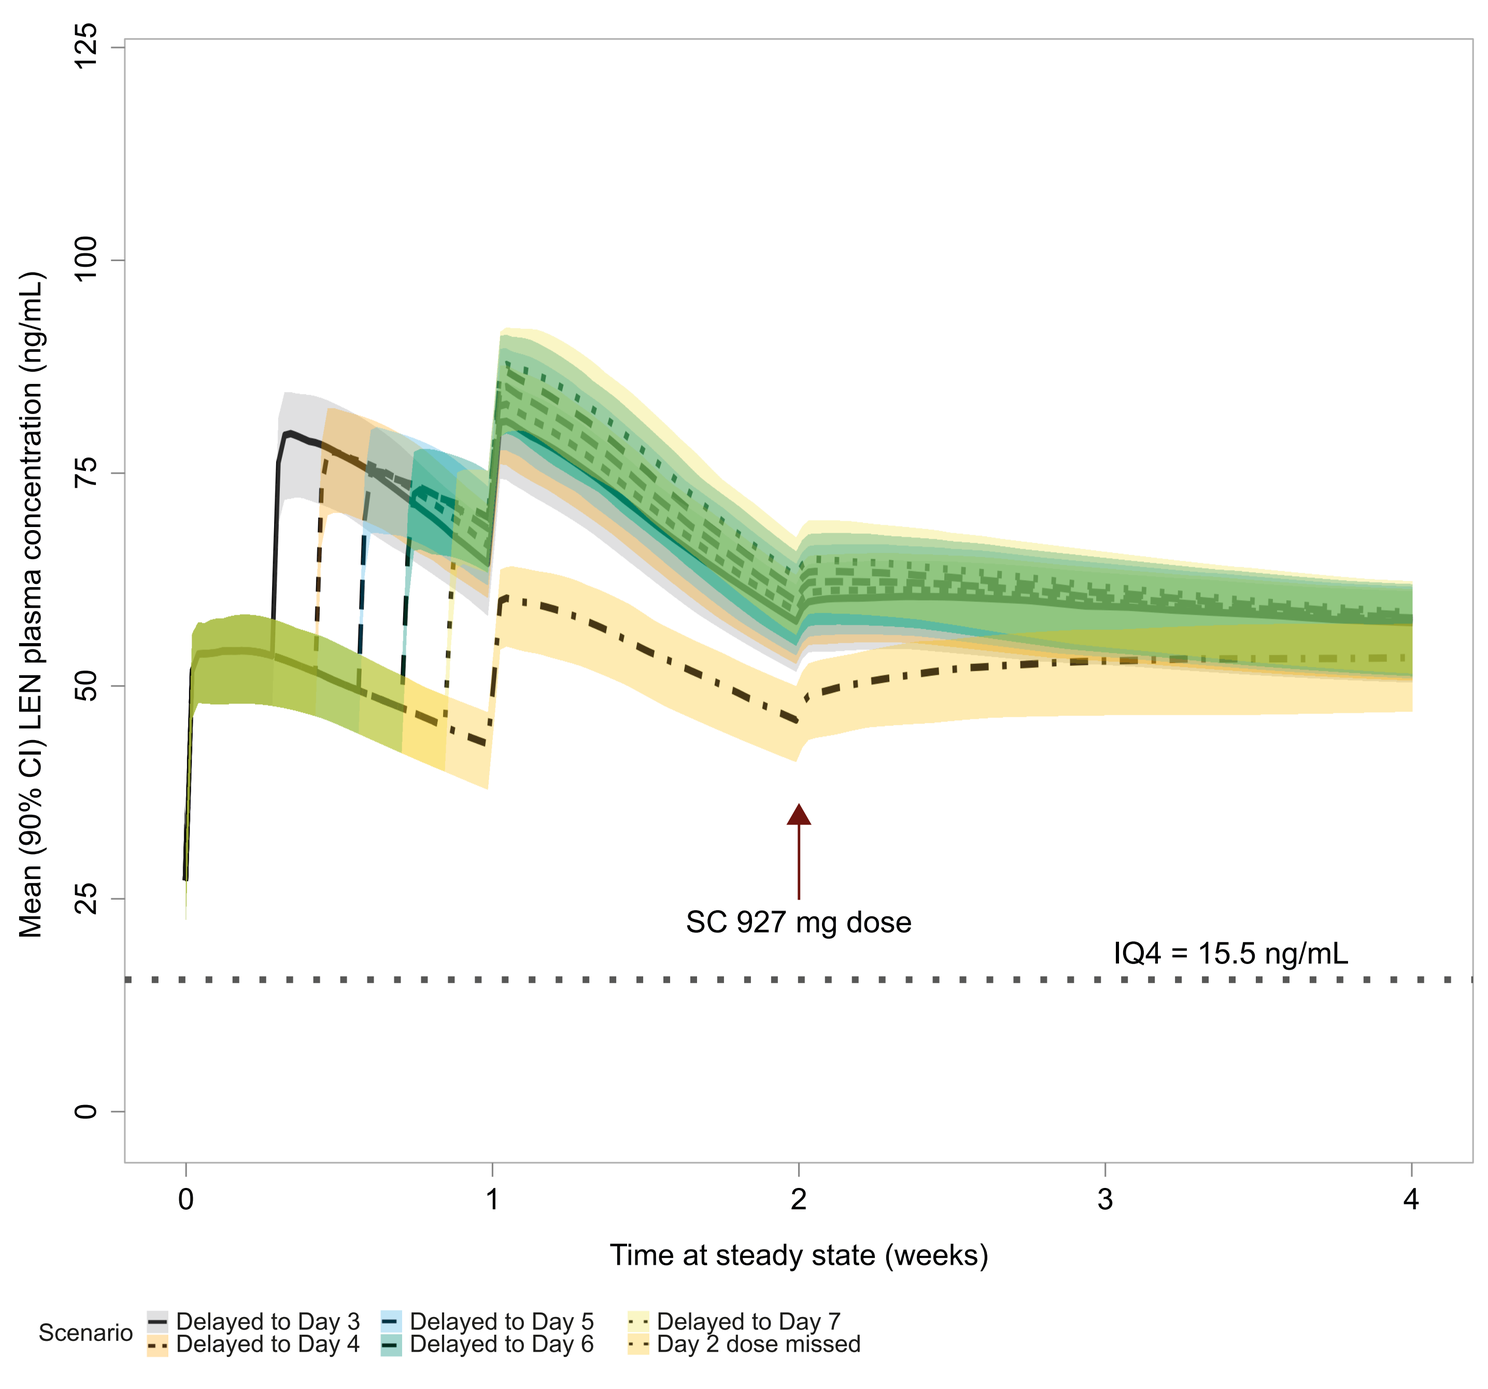
Mean (90% CI) LEN plasma concentration (ng/mL)
SC 927 mg dose
IQ4 = 15.5 ng/mL
Time at steady state (weeks)
Scenario
Delayed to Day 3
Delayed to Day 4
Delayed to Day 5
Delayed to Day 6
Delayed to Day 7
Day 2 dose missed

B) Day 2 oral dose missed by ≥6 days

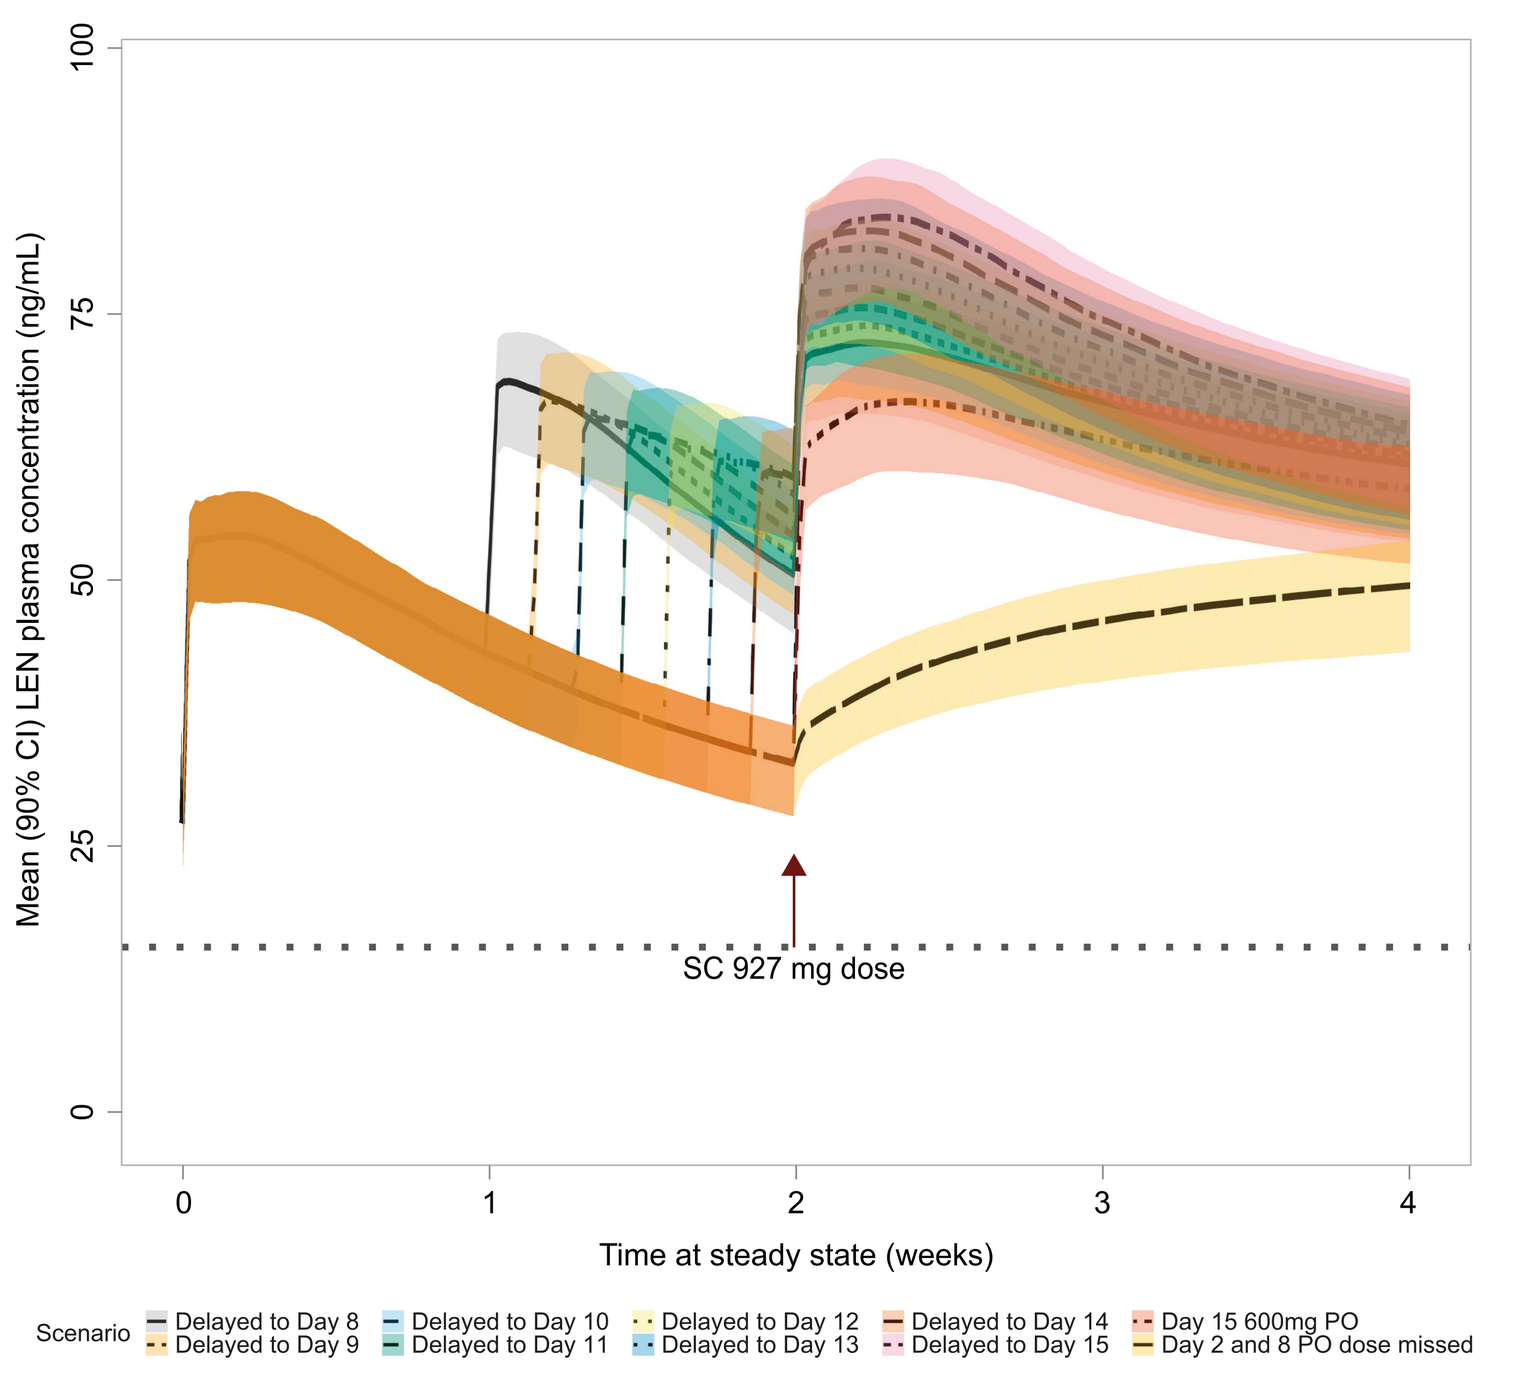
Mean (90% CI) LEN plasma concentration (ng/mL)
SC 927 mg dose
Time at steady state (weeks)
Scenario
Delayed to Day 8
Delayed to Day 9
Delayed to Day 10
Delayed to Day 11
Delayed to Day 12
Delayed to Day 13
Delayed to Day 14
Delayed to Day 15
Day 15 600mg PO
Day 2 and 8 PO dose missed

C) Day 8 oral dose missed by <6 days

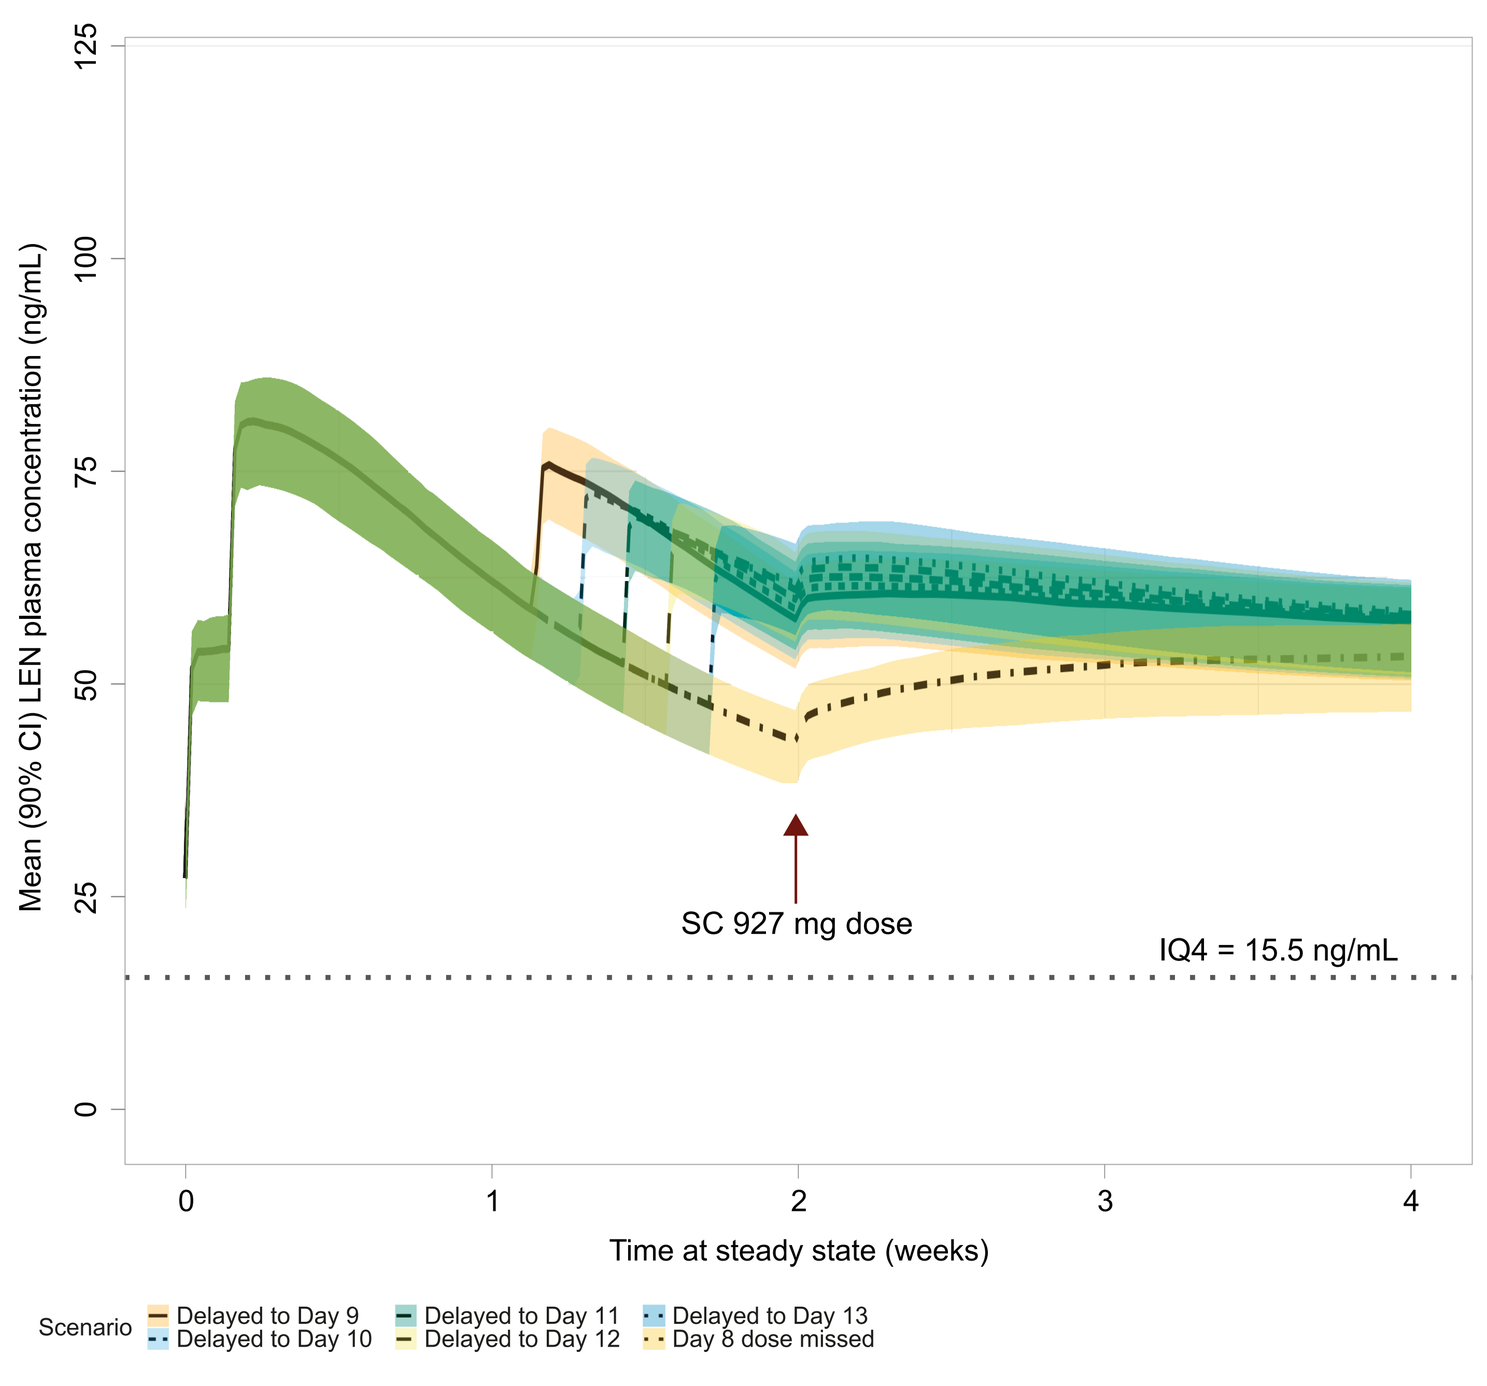
Mean (90% CI) LEN plasma concentration (ng/mL)
SC 927 mg dose
IQ4 = 15.5 ng/mL
Time at steady state (weeks)
Scenario
Delayed to Day 9
Delayed to Day 10
Delayed to Day 11
Delayed to Day 12
Delayed to Day 13
Day 8 dose missed

D) Day 8 oral dose missed by ≥6 days

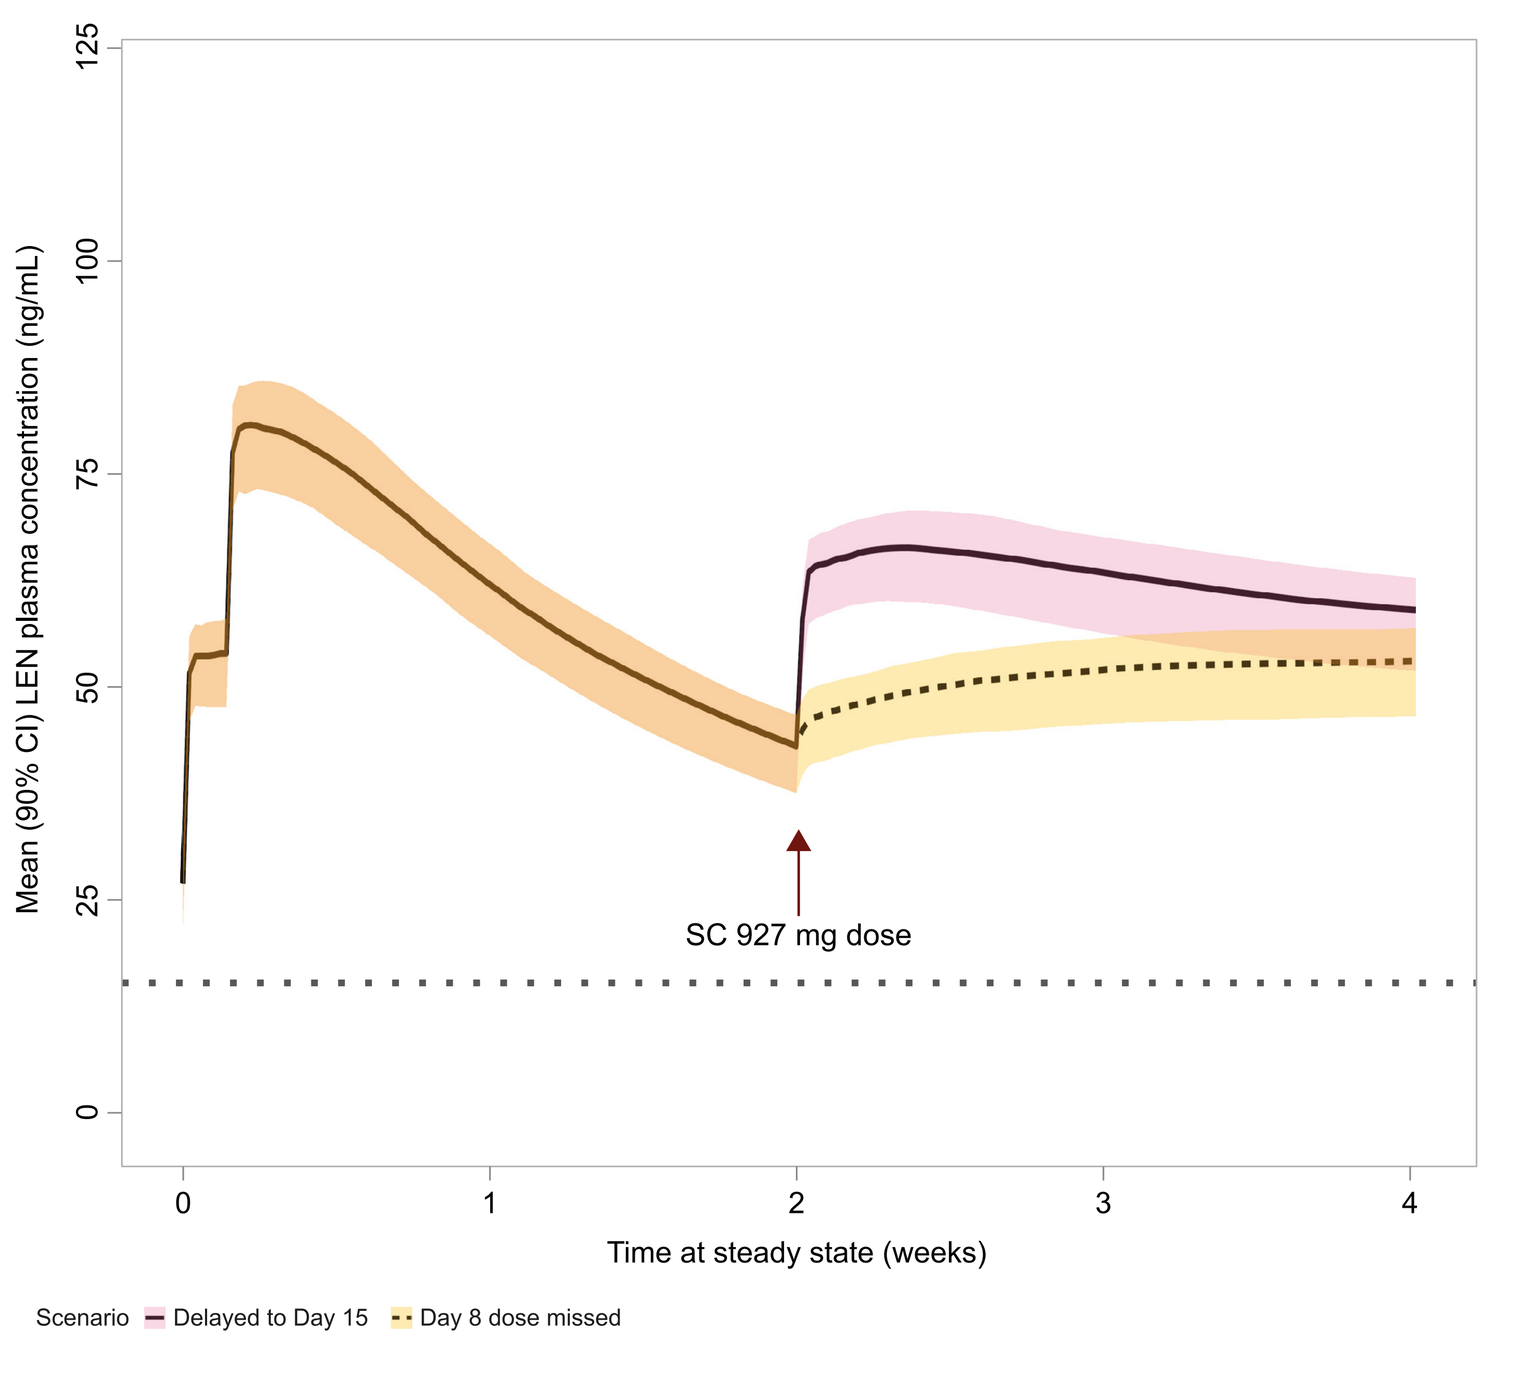
Mean (90% CI) LEN plasma concentration (ng/mL)
SC 927 mg dose
Time at steady state (weeks)
Scenario
Delayed to Day 15
Day 8 dose missed

Lines represent the simulated mean. Shaded areas represent the 90% confidence interval of the simulated mean. In all scenarios, LEN SC dose was simulated on Day 15. Simulations for restart of treatment were conducted at steady state with dosing at 28 weeks after last SC injection.
CI, confidence interval; IQ4, inhibitory quotient-4; LEN, lenacapavir; PO, per os (oral); PWH, people with HIV; SC, subcutaneous

References: 1. Sunlenca US PI 2022, available on https://www.accessdata.fda.gov/drugsatfda_docs/label/2022/215973s000lbl.pdf last accessed 10 July 2023; 2. Sunlenca EU SmPC 2022, available on https://www.ema.europa.eu/en/documents/product-information/sunlenca-epar-product-information_en.pdf last accessed 10 July 2023; 3. Segal-Maurer S, et al. *N Engl J Med* 2022;386:1793–1803; 4. Shaik N, et al. Poster PESUB23 presented at AIDS 2022; 5. Link JO, et al. *Nature* 2020;584:614–618;6. Shaik N, et al. Poster EPB174 presented at AIDS 2022

Acknowledgments: These analyses were conducted by Certara, Inc., and were funded by Gilead Sciences, Inc. Medical writing support was provided by Jackie Phillipson of Ashfield MedComms (Macclesfield, UK), an Inizio company, and funded by Gilead Sciences, Inc.

Disclosures: **R Singh**, **N A Shaik**, **M Rhee**, and **R Palaparthy** are all employees and shareholders of Gilead Sciences, Inc. **F Bellanti**, **C Comisar**, and **K Polireddy** are all employees and shareholders of Certara, Inc.

Presented at the 12th IAS Conference on HIV Science, 23–26 July 2023, Brisbane, Australia